FOL-005 – modulation of hair growth in clinical studies

Jan Alenfall[1], Maria Ekblad[1], Jan Kottner[2], Annika Vogt[2], Ulrike Blume-Peytavi[2]. [1)] Follicum AB, Lund, Sweden, [2)] Charité-Universitätsmedizin Berlin, Germany

Introduction and Objective

Hair loss comes in a wide range of clinical manifestations with androgenetic alopecia being the most frequently observed entity, affecting up to 80% of men and 50% of women[1]. Even though this disorder is known to cause substantial psychological distress to many patients, therapeutic options are currently limited. The multitude of signalling pathways involved in hair growth suggests that different classes of molecules could be successful candidates for hair growth stimulation. Still, the number of active pharmaceutical candidates reaching convincing evidence in clinical trials is low. New pharmacological treatment strategies that offers safe and effective agents for the treatment of hair-loss need to be developed.

FOL-005 is a well-characterized osteopontin derived peptide that binds specifically to hair follicles (HFs). Osteopontin, a multifunctional immunomodulatory glycoprotein, has been shown to exert multiple roles in skin physiology and pathology[2, 3, 4] and is significantly expressed in human HFs[5]. The hypothesis that osteopontin-derived fragments may modulate human hair growth, has been tested in several preclinical studies with positive results and FOL-005 has undergone a standard battery of toxicological tests without signs of any overt toxicity.

Two clinical studies with FOL-005 have been completed. In the first study, "*Investigation of FOL-005 on Clinical Safety and Effect on Hair Growth*", FOL-005 at different doses was injected into the forefront of the thighs of healthy men, two or three times a week for three months. The results showed that the treatment was safe and a significant increase in hair density was shown at one of the doses.

The second study, "*A randomised, double-blind, placebo-controlled phase 2 trial of FOL-005 to investigate efficacy on hair growth on scalp skin in healthy volunteers*", was recently completed. In this study the efficacy and safety of FOL-005 administered as intradermal injections into the scalp, three times per week for three months, was studied. The study confirmed an excellent safety profile and although no significant results on hair density were obtained, several interesting effects on the secondary endpoints (hair growth parameters) suggest a hair growth promoting effect of FOL-005. Further studies are needed to investigate hair growth promotion capacities of FOL-005 in a new topical formulation where more frequent applications can be made.

In parallel with the clinical development, a novel formulation for topical use has been developed. The formulation was tested for efficacy in an *in vivo* experimental mouse model. Currently a clinical phase II study with the topical formulation is planned.

Experimental

Clinical phase I/IIa study "Investigation of FOL-005 on Clinical Safety and Effect on Hair Growth"

The clinical trial was a single centre, randomized, double-blind, placebo-controlled safety trial. In order to study several different treatments on the same subject, the study was performed on six separate areas on the forefront of the thighs of healthy volunteers. The study was divided in two parts, a single ascending dose (SAD) part, and a multiple dose (MD) regime part. Five of the areas were treated with either placebo or one of four doses of FOL-005 in a double-blinded manner and one area was left untreated. FOL-005 and placebo were administered as intradermal injections. The subjects were monitored continuously for safety signals during the study. The number of hairs at the separate areas were manually counted according to a standardized procedure.

Clinical phase IIa study "A randomised, double-blind, placebo-controlled phase 2 trial of FOL-005 to investigate efficacy on hair growth on scalp skin in healthy volunteers"

The Phase II trial was a randomized, double-blind, placebo-controlled safety and efficacy trial. FOL-005 was administered as intradermal injections to minizones on the scalp of men with androgenic alopecia of grade 3V to 4/4a according to Norwood/Hamilton. Two minizones on the scalp of each subject were identified and each one of the separate areas were treated with either of four different active concentrations or placebo. The subjects were treated three times a week and were monitored continuously for safety signals during the study. The hair growth potential was assessed by using TrichoScan® software according to a standardized procedure.

In vivo efficacy study of topical formulation in C57Bl/6 mice

The hair growth stimulation properties were investigated in young C57Bl/6 mice in stable telogen phase of hair growth. The fur at the back of the animals was carefully clipped and treated with a topical formulation of FOL-005 or placebo, once daily. In addition one group was administered Minoxidil 5% (highest dose commercially available) twice daily. The hair growth was visually inspected and scored on a daily basis.

Results

Investigation of FOL-005 on Clinical Safety and Effect on Hair Growth – a phase I/IIa study

CHARITÉ UNIVERSITÄTSMEDIZIN BERLIN

- Intradermal injections on the thighs
- Administrations controlled and done at clinic
- All subjects received all doses
- Two groups, 2 or 3 times weekly for 3 months
- 3 months follow-up

Multiple Dosing
30 volunteers, 3 months

A: 2x weekly

B: 3x weekly

Six Treatment Areas
1. No Treatment
2. Placebo
3. Dose 1
4. Dose 2
5. Dose 3
6. Dose 4

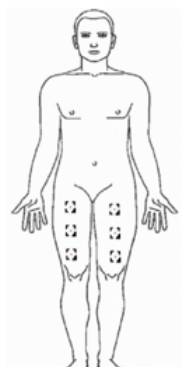

Hair density was assessed prior to and after 13 weeks of treatment by photography and manually hair counting by two separate investigators.

Placebo		FOL-005	
Baseline	13 weeks	Baseline	13 weeks

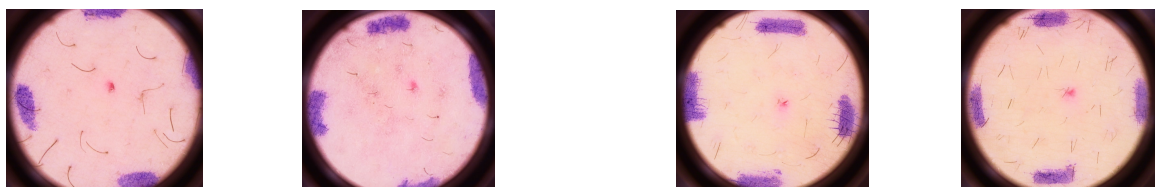

*Fotofinder Device, 20x magnification

Example photographs, hair count before and after 13 weeks of treatment

The hair growth stimulatory effect was assessed by counting the number of hairs prior to treatment and after 13 weeks of treatment.

- **A significant increase in hair density of +8% (p=0.016) was observed at Dose 2 of FOL-005**
- **Placebo showed a decrease of -5% (p=0.513)**
- **In some subjects, FOL-005 treatment induced up to 60% increase in hair density compared to baseline**

Unique topical formulation of FOL-005 potently increased hair growth in mouse model

- Established hair growth model
- C57Bl mice in stable telogen phase,7 to 9 weeks old
- Fur at the back carefully clipped
- Three different strengths of FOL-005 or placebo, once daily
- Minoxidil highest commercial strength, twice daily

Hair growth scoring and grading criteria

Observation	Score
No hair growth, pink skin	0
Skin colour change from pink to grey without visible hair growth	0.5
Skin colour change from pink to grey or black, without visible hair growth, indicating onset of anagen	1.0
Sparse hair growth	1.5
Sparse or diffuse short hair growth	2.0
Moderate hair growth	2.5
Dense, normal coat	3.0

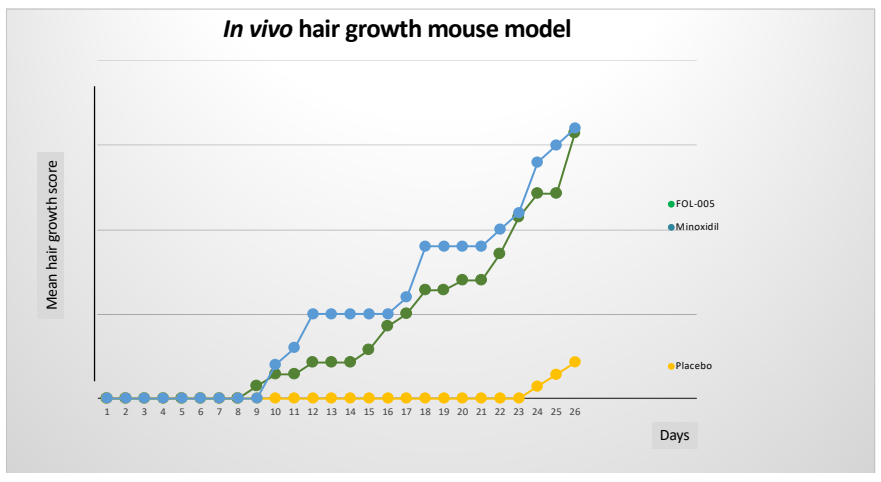
In vivo hair growth mouse model
Mean hair growth score
FOL-005
Minoxidil
Placebo
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26
Days

Hair growth score after treatment with FOL-005 at different doses or placebo once daily. Minoxidil twice daily for comparison

- **Hair growth was initiated after 1 week of treatment**
- **FOL-005 effect showed a dose-response**
- **Effect of FOL-005 comparable to Minoxidil**

A randomised, double-blind, placebo-controlled phase 2 trial of FOL-005 to investigate efficacy on hair growth on scalp skin in healthy volunteers

CHARITÉ UNIVERSITÄTSMEDIZIN BERLIN

bioskin® unique in dermatology research

Parameter	Description
Treatment area	Mini zones in hair/non hair borderline area on scalp, 2 zones/subject
Subjects	Alopecia patients with Norwood/Hamilton grade 3V to 4/4a
Number of subjects	60
Number of doses	Four active doses and placebo
Number of areas/dose	Active doses 25, placebo 20
Administration	Intradermal injections 50 µL
Treatment frequency	Three times a week
Treatment duration	3 months
Efficacy endpoints	Hair growth parameters assessed by TrichoScan® software, hair density, anagen hair density, telogen hair density, hair thickness, hair length, hair growth rate etc

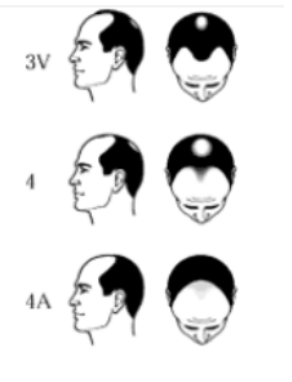
3V
4
4A

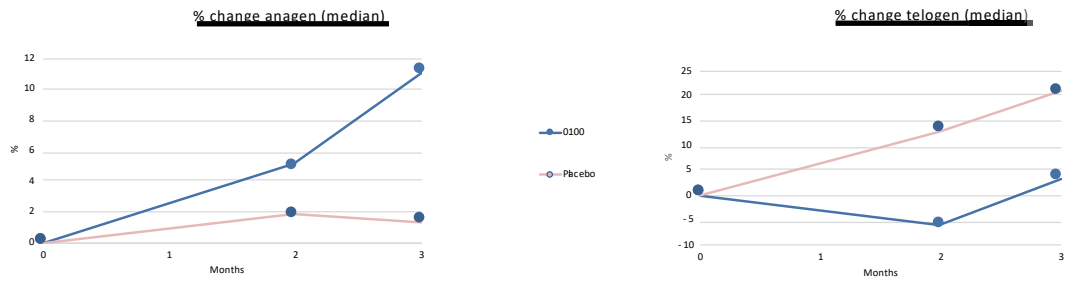
% change anagen (median)
% change telogen (median)
%
Months
0100
Placebo

Changes in percentage of hair follicles in anagen and telogen phase respectively after treatment for 3 months with either of four doses of FOL-005 or placebo, as compared to prior to treatment.

Primary endpoint
Hair density increased with 7 hairs/cm^2 (p=0.0782)

Selected secondary endpoints
FOL-005: Increase in percentage of hair follicles in anagen phase (p=0.254)
Placebo: Increase in percentage of hair follicles in telogen phase (p=0.0288)

Conclusions

Clinical studies

From the results of the two Phase 2a clinical studies with FOL-005 it was concluded that the compound was very well tolerated. A tendency towards increased hair density was seen in both studies. In addition interesting signals on several secondary hair growth parameters were obtained.

***In vivo* mouse model with new topical formulation**

A hair-growth promoting effect was demonstrated after treatment with a novel topical formulation of FOL-005.

Forthcoming plans

Further proof of concept studies are needed to investigate the full hair growth promotion capacities of FOL-005 with the unique and newly developed topical formulation, hence a phase 2a study with the topical formulation is currently planned with higher and optimised dosage.

About Follicum

Follicum is a Swedish biotechnology company working with development of new peptide pharmaceuticals to control hair growth and to treat diabetes and its complications.

If you would like to discuss collaboration with Follicum please contact:

Jan Alenfall, PhD (CEO). Phone: +46 709 315 115. Email: jan.alenfall@follicum.com Address: Follicum AB, Scheelevägen 22, SE-223 63 Lund, Sweden Web: www.follicum.com

References: [1]Kelly et al., 2016; [2]Kothari et al., 2016; [3]Scatena et al., 2007; [4]Wei et al., 2017; [5]Chang et al., 2008